Phase II Trial of BC-819 Intravesical Gene Therapy in Combination with BCG in Patients with Non-Muscle Invasive Bladder Cancer (NMIBC).

Sarel Halachmi, Ilan Leibovitch, Amnon Zisman, Avi Stein, Shalva Benjamin, Ami Sidi, Ron Knickerbocker, Michal Limor, Yan Moore; Bnai Zion Medical Center, Haifa, Israel; Meir Medical Center, Kfar Saba, Israel; Shamir Medical Center, Tzrifin, Israel; Carmel Medical Center, Haifa, Israel; Edith Wolfson Medical Center, Holon, Israel; BioCancell, Cambridge, MA.

Background:

BC-819 is an intravesical gene therapy consisting of a recombinant DNA construct which delivers a lethal cellular toxin, engineered from diphtheria, specifically to malignant cells in the bladder. In this study we tested the feasibility of administering BC-819 in combination with BCG with the goal of improving outcomes for patients with NMIBC.

Methods:

This phase II multi-center trial assessed 3 intravesical combination induction schedules of BC-819 and BCG; alternating (A), sequential (S), and twice weekly (TW) for 6-12 weeks. The population included adult patients for whom BCG was indicated. All patients underwent transurethral resection of the bladder tumor (TURBT) prior to study entry. The primary endpoint was safety; recurrence was assessed as a secondary endpoint.

Results:

38 Caucasian patients were included in the intent-to-treat population; 16 in A, 6 in S, and 16 in TW cohorts. The median follow-up is 18-mo. The mean age was 69 years (±11.3), 92% were males. 29% had prior BCG. 16% of patients had CIS lesions, and 47% had multiple recurrences prior to participation. In 86% the resected tumor size ≥1.0cm. The overall median time to recurrence (mTTR) was not reached. The 24-months rates of recurrence and progression were 46% and 24%, respectively. The longest median time to recurrence among the three schedules was in the alternating group, but due to the small numbers of patients in each group the best schedule could not be determined. Therapy was associated with 5 AEs (in 5 patients; 13%) related to BC-819 (2 UTIs – 1 mild/1 moderate, 2 hematurias – both mild, 1 dysuria - mild) and 29 related to BCG. 3 SAEs were reported, none related to BC-819.

The Drug

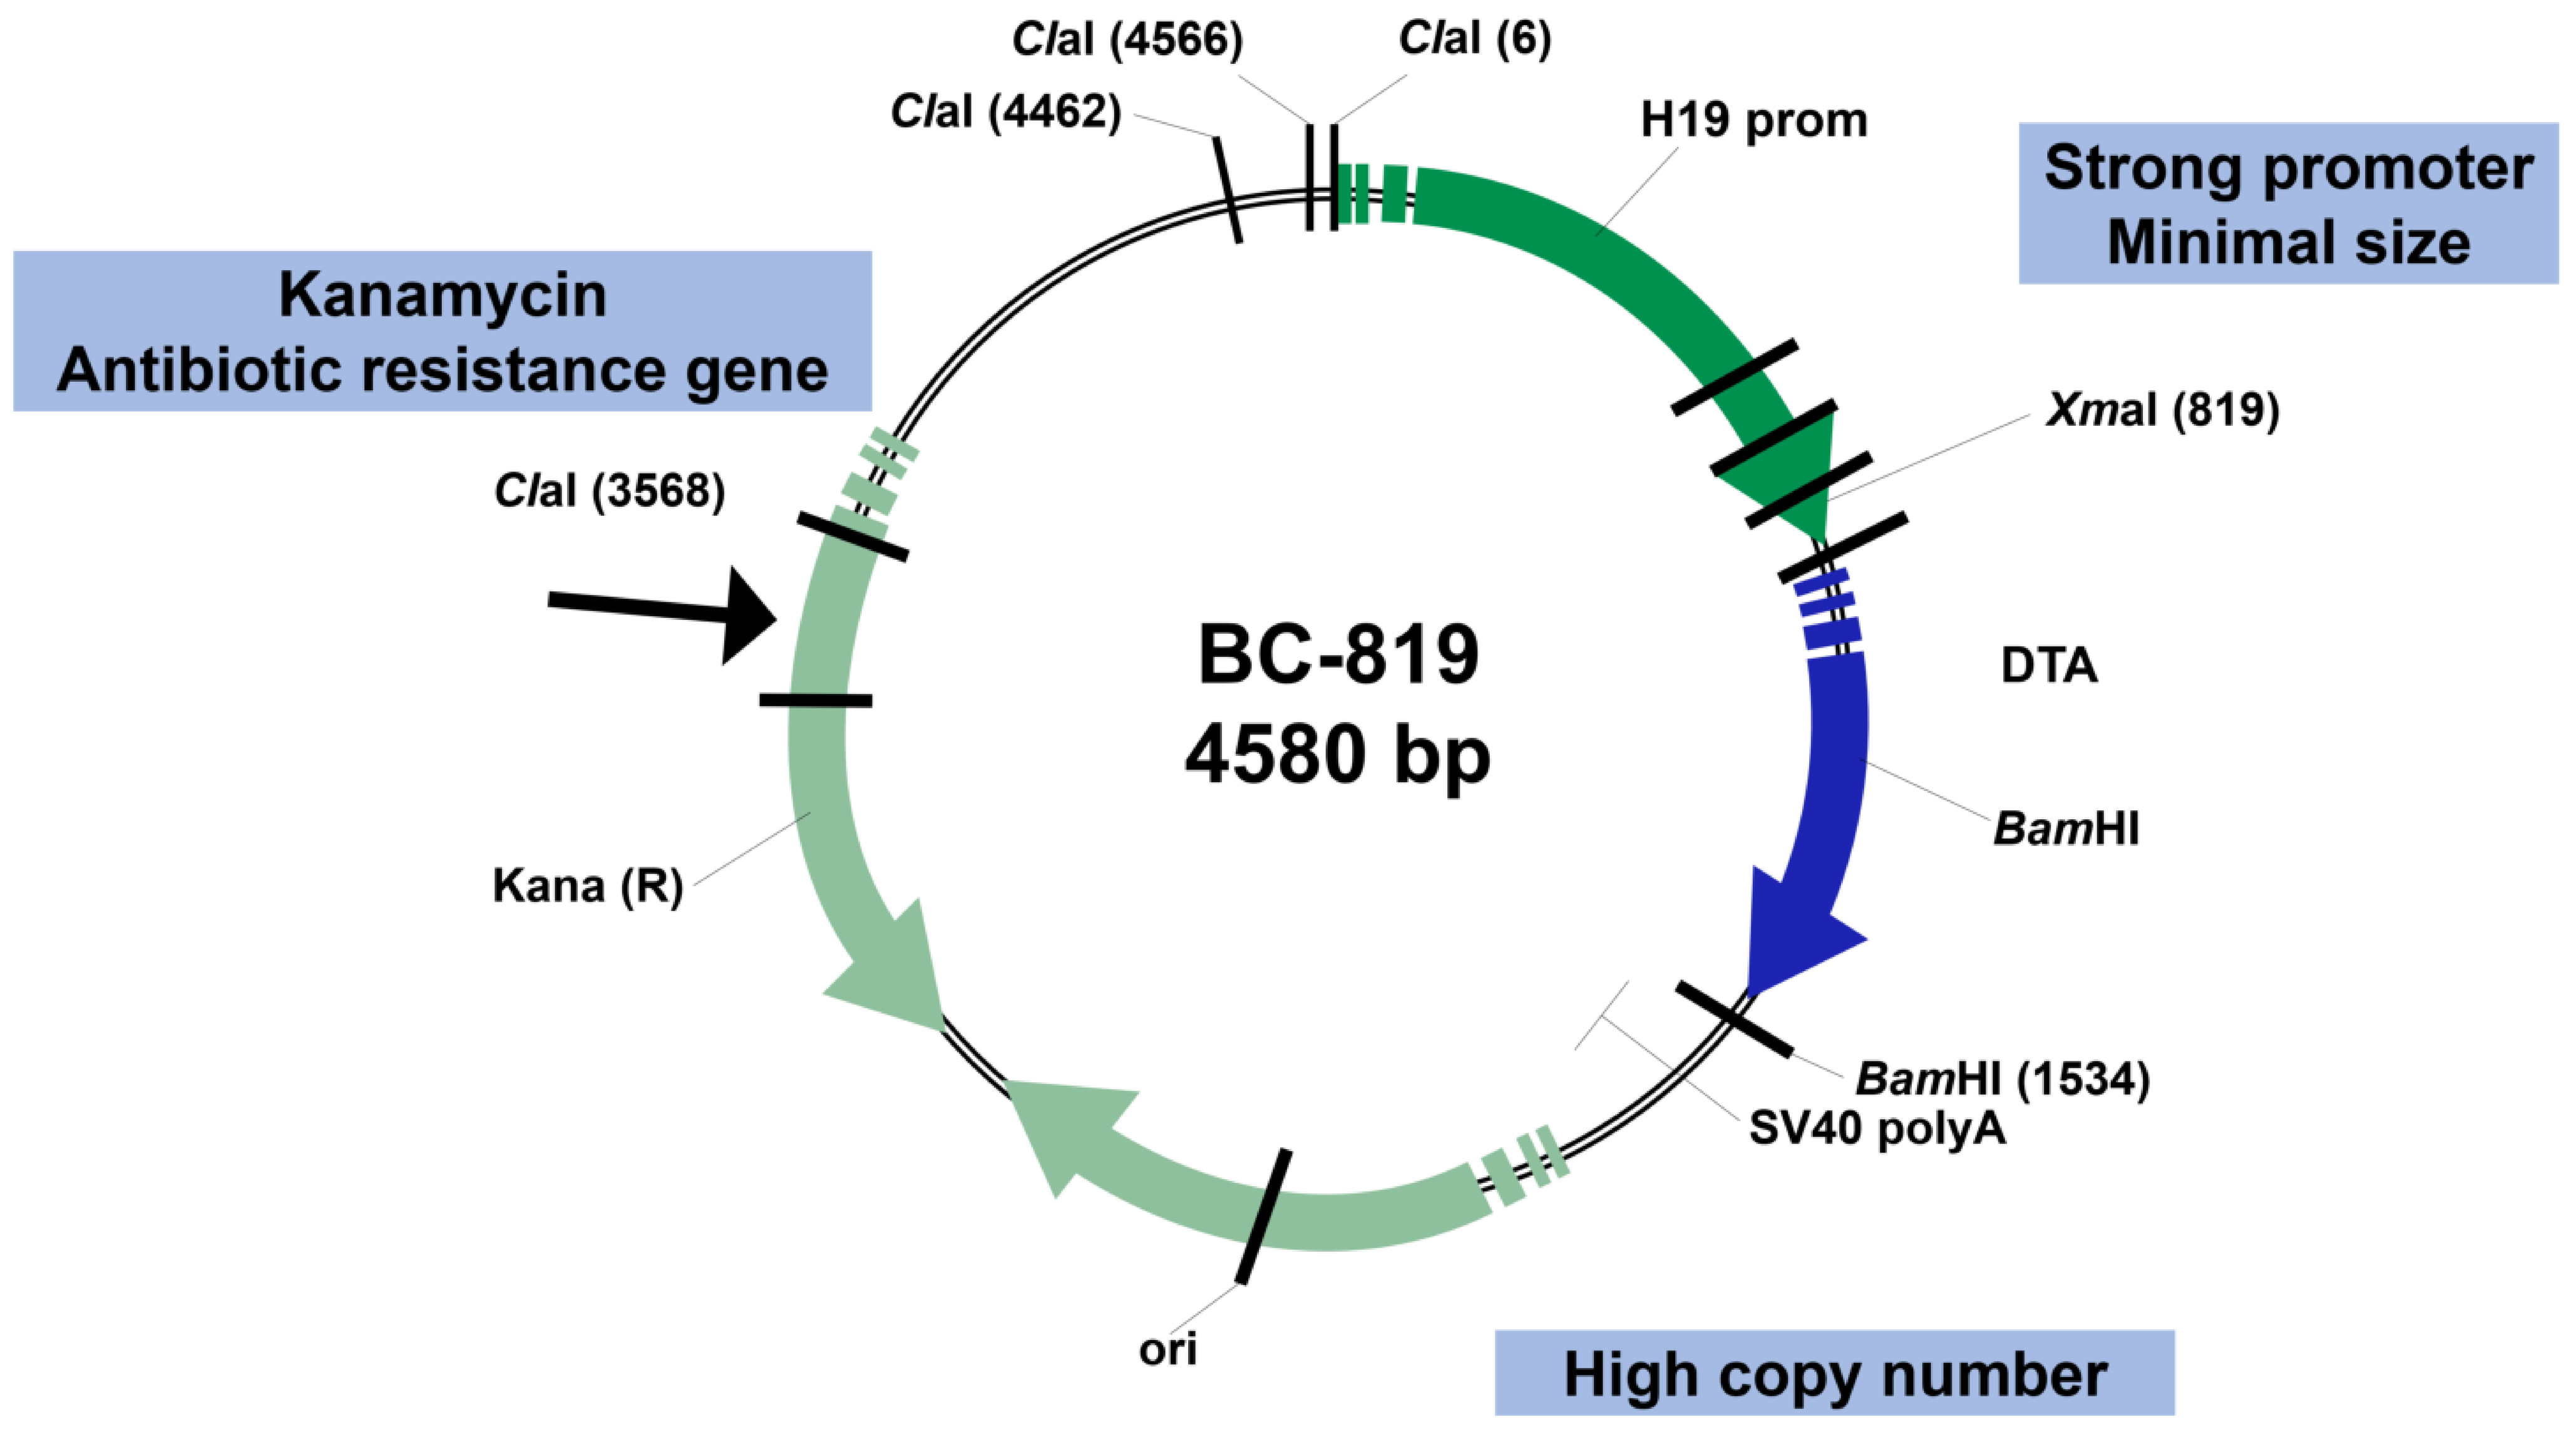
ClaI (4566)
ClaI (6)
ClaI (4462)
H19 prom
Strong promoter
Minimal size
Kanamycin
Antibiotic resistance gene
XmaI (819)
ClaI (3568)
BC-819
4580 bp
DTA
BamHI
Kana (R)
BamHI (1534)
SV40 polyA
ori
High copy number

Road Map of Cancer
The Oncogene RNA H19 in Human Cancer

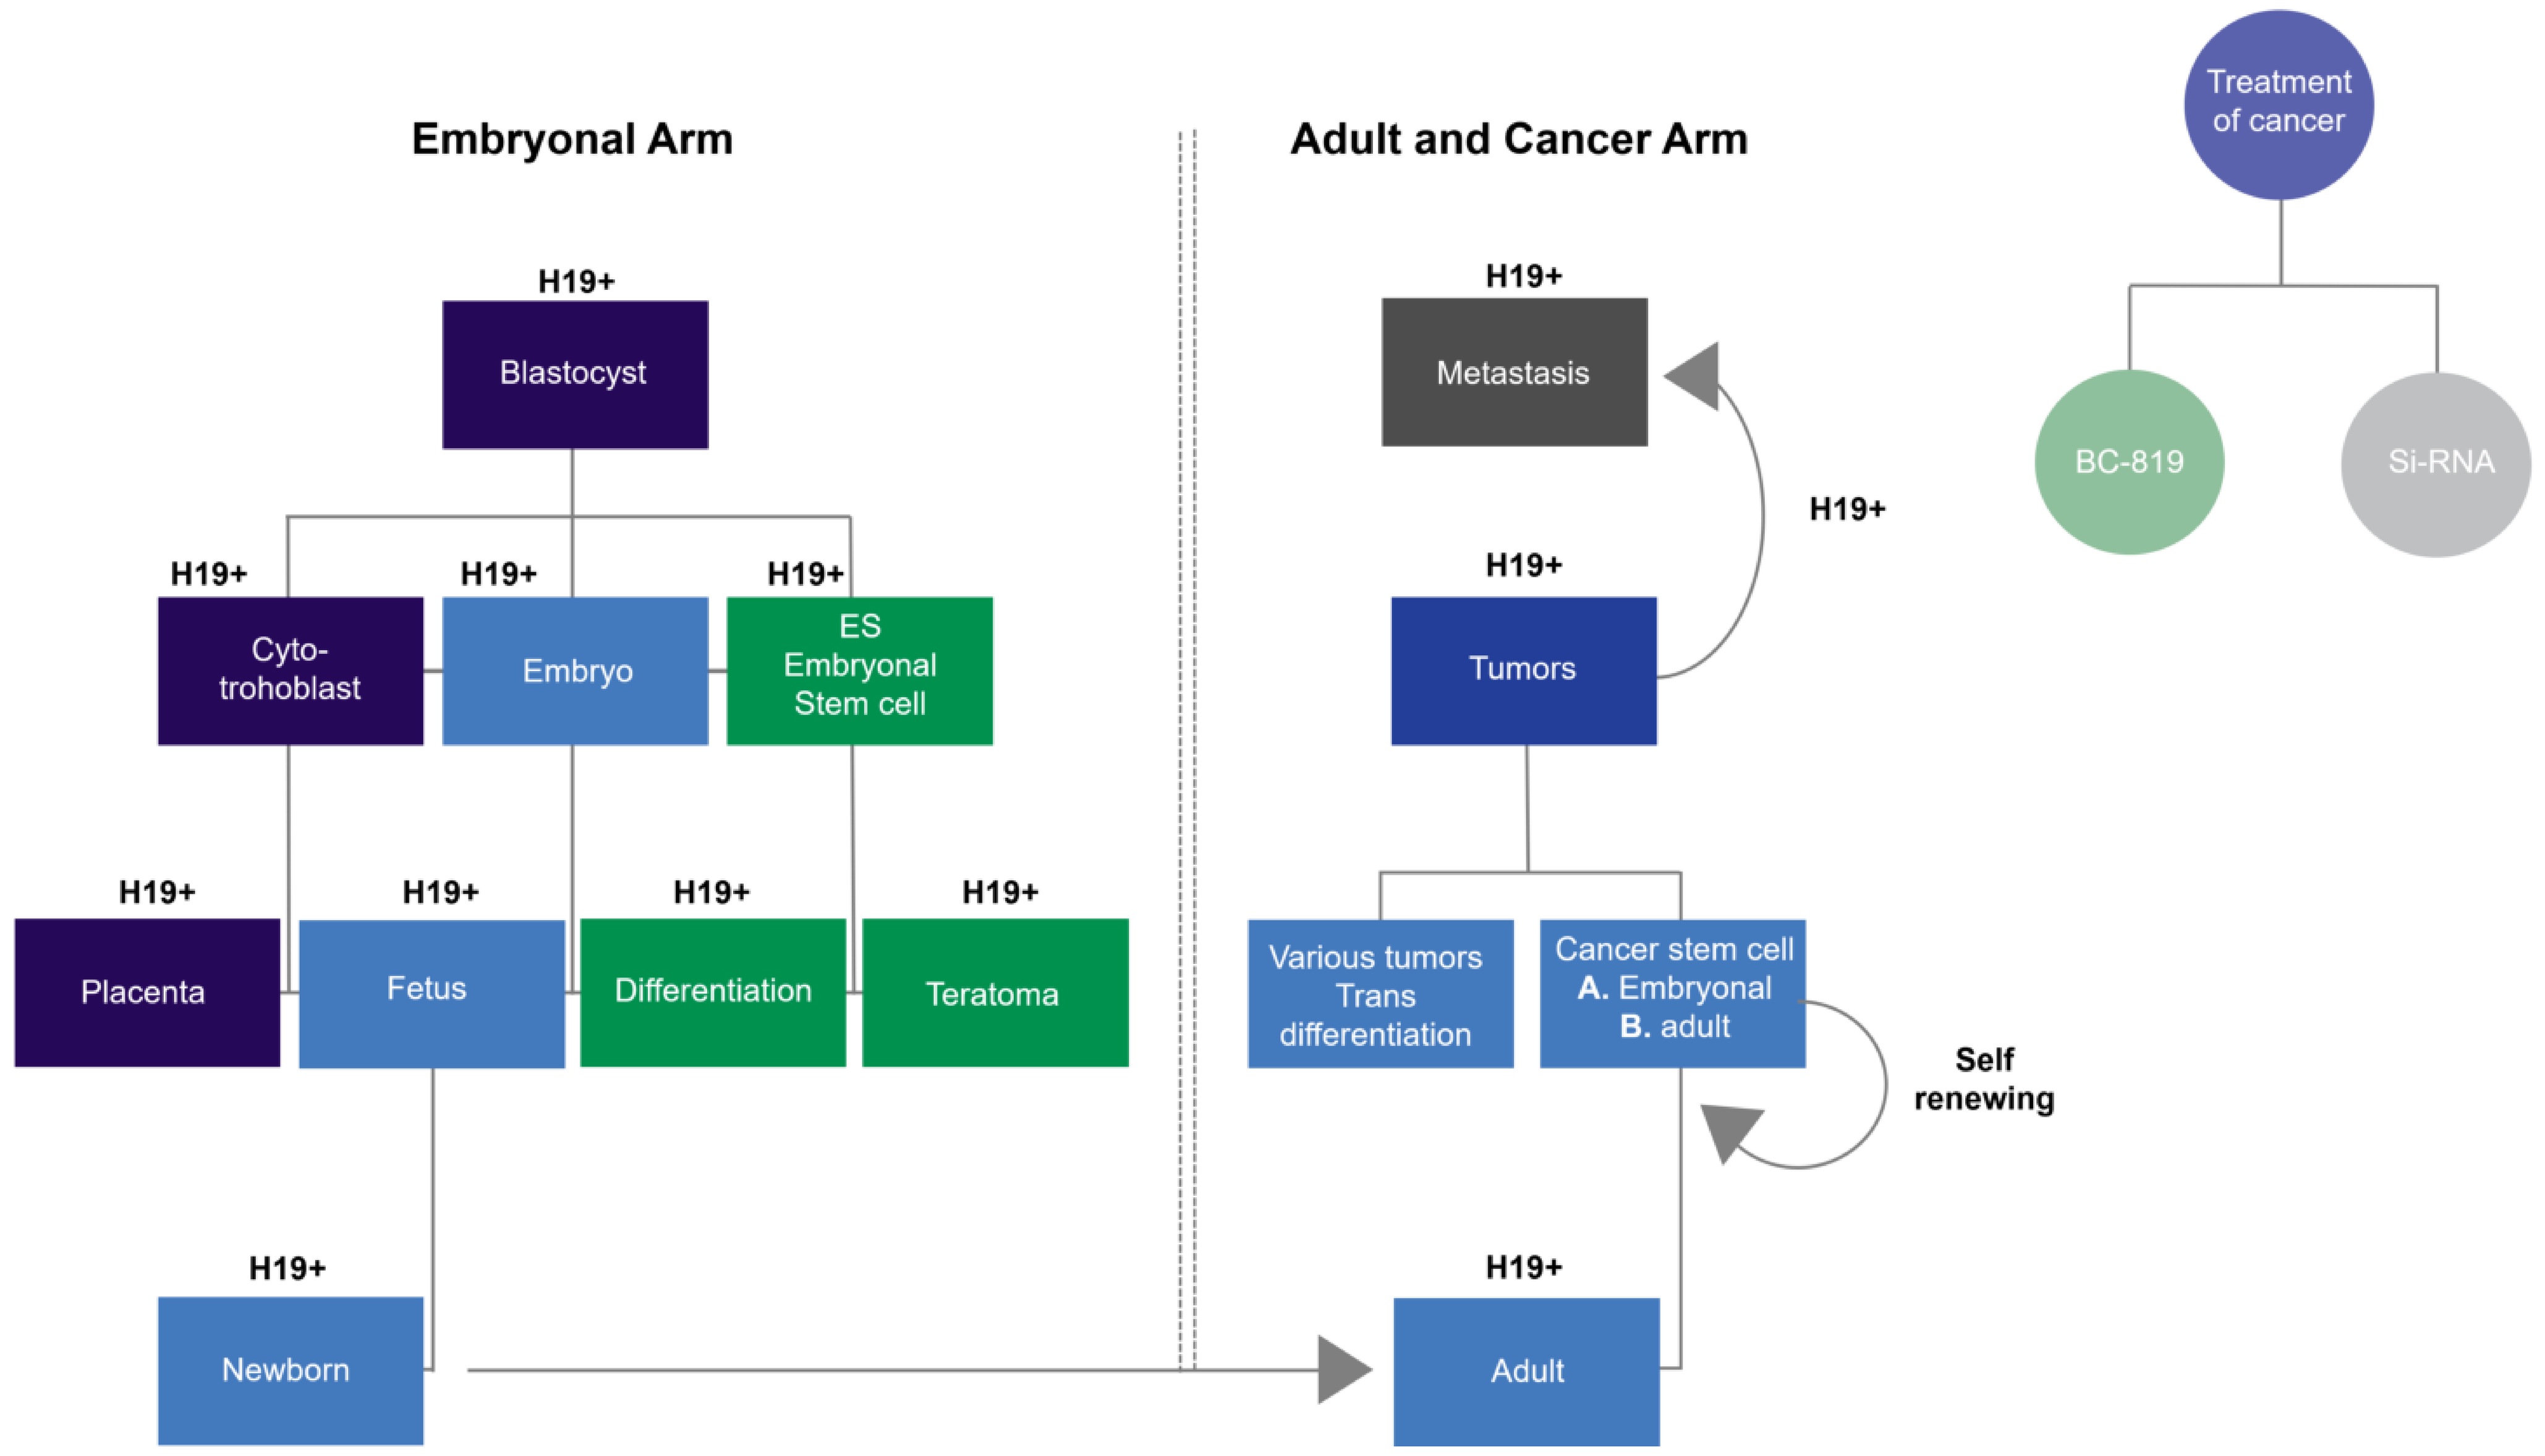
Embryonal Arm
Adult and Cancer Arm
Treatment of cancer
H19+
Blastocyst
Cyto-trohoblast
Embryo
ES Embryonal Stem cell
Placenta
Fetus
Differentiation
Teratoma
Newborn
Metastasis
Tumors
Various tumors Trans differentiation
Cancer stem cell A. Embryonal B. adult
Self renewing
Adult
BC-819
Si-RNA

Month	Recurrence Free Survival	Progression Free Survival
3	94.6	100.0
6	78.4	94.6
9	73.0	89.2
12	67.6	89.2
18	62.2	83.8
Overall (24-mo)	54.1	75.7

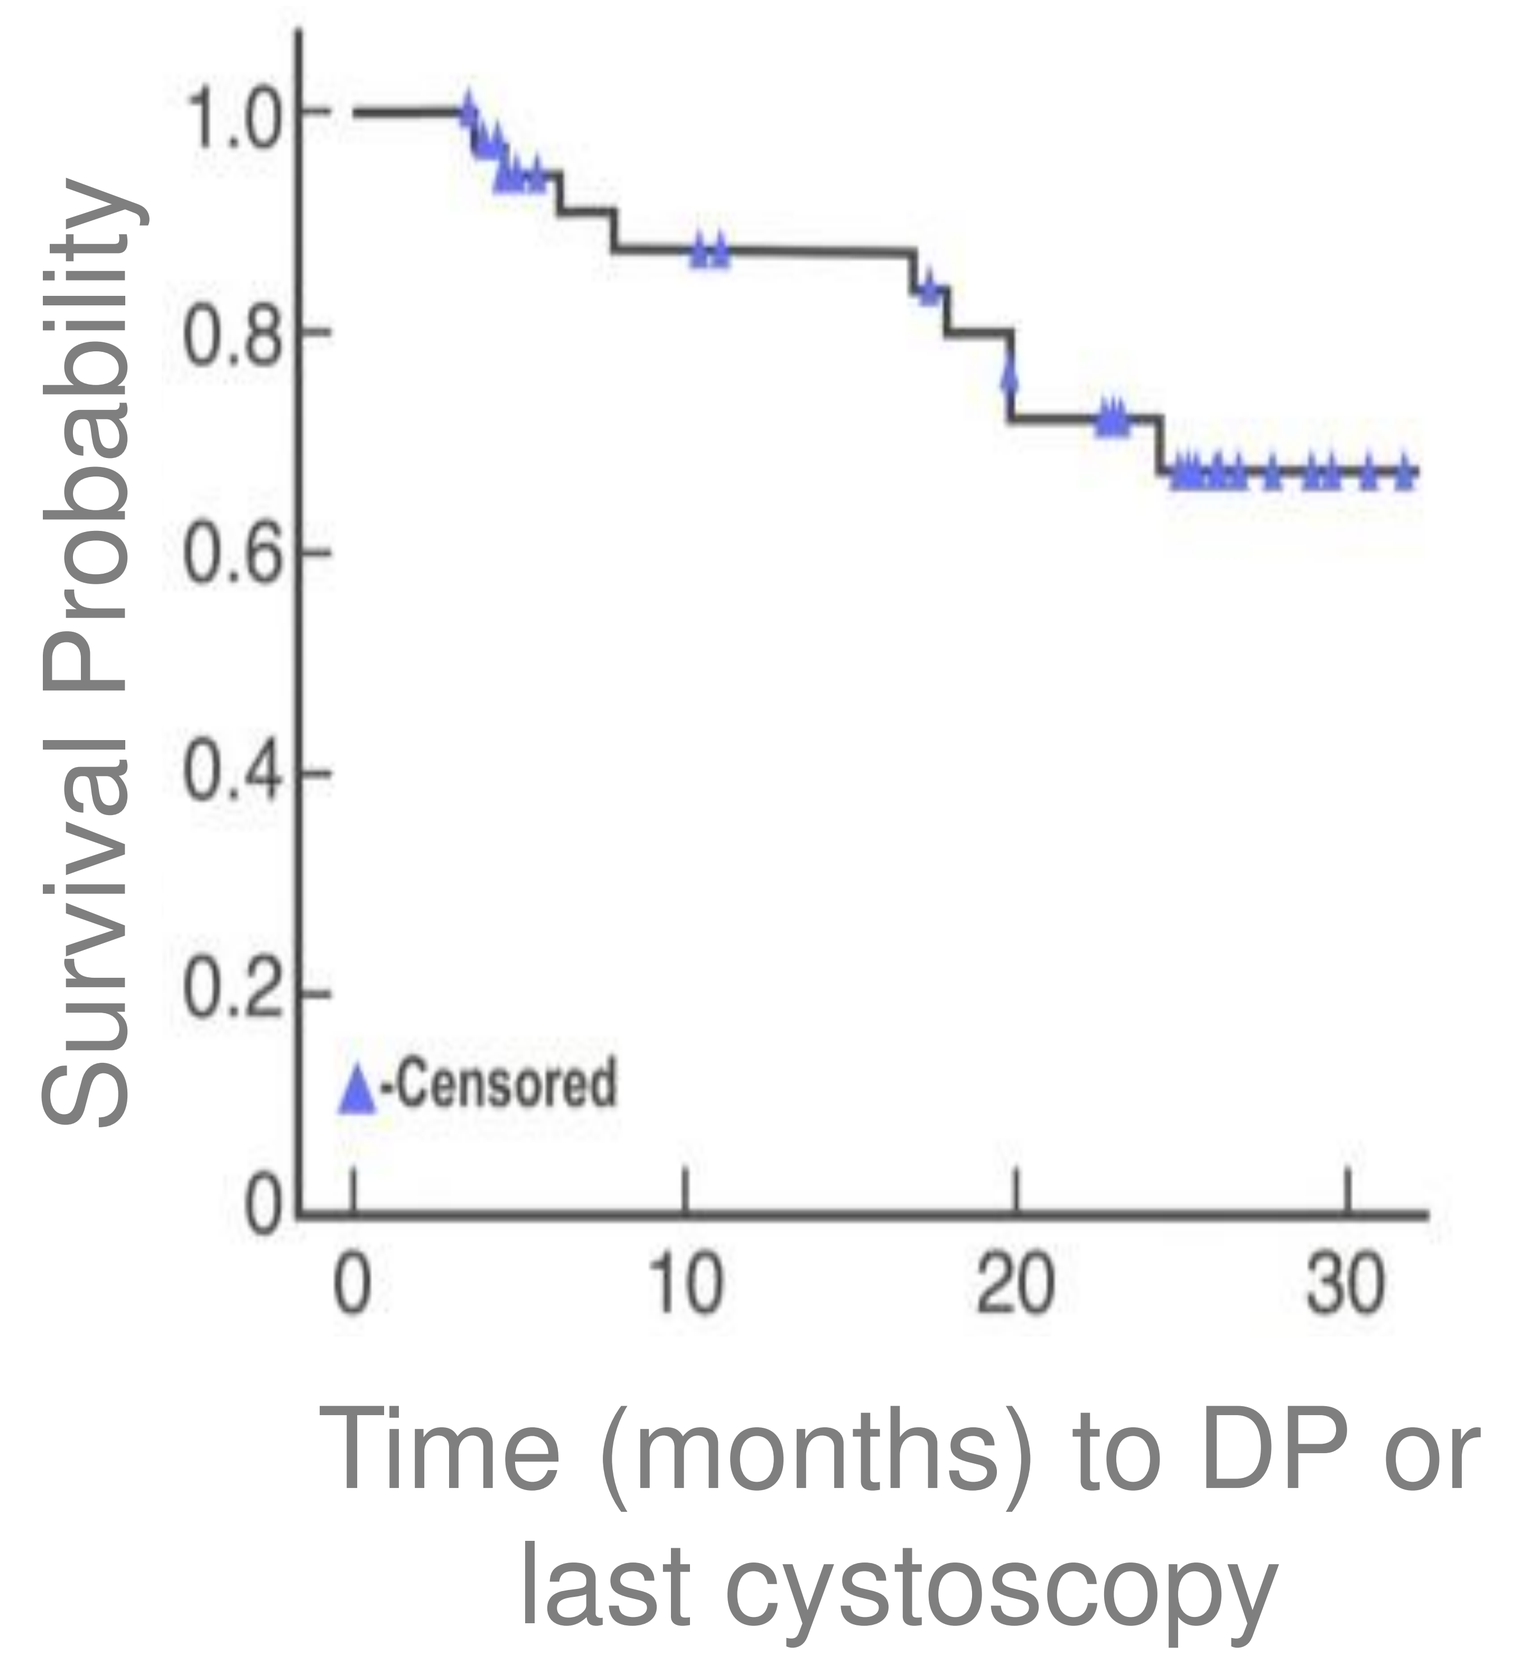
Survival Probability
1.0
0.8
0.6
0.4
0.2
0
-Censored
0
10
20
30
Time (months) to DP or last cystoscopy

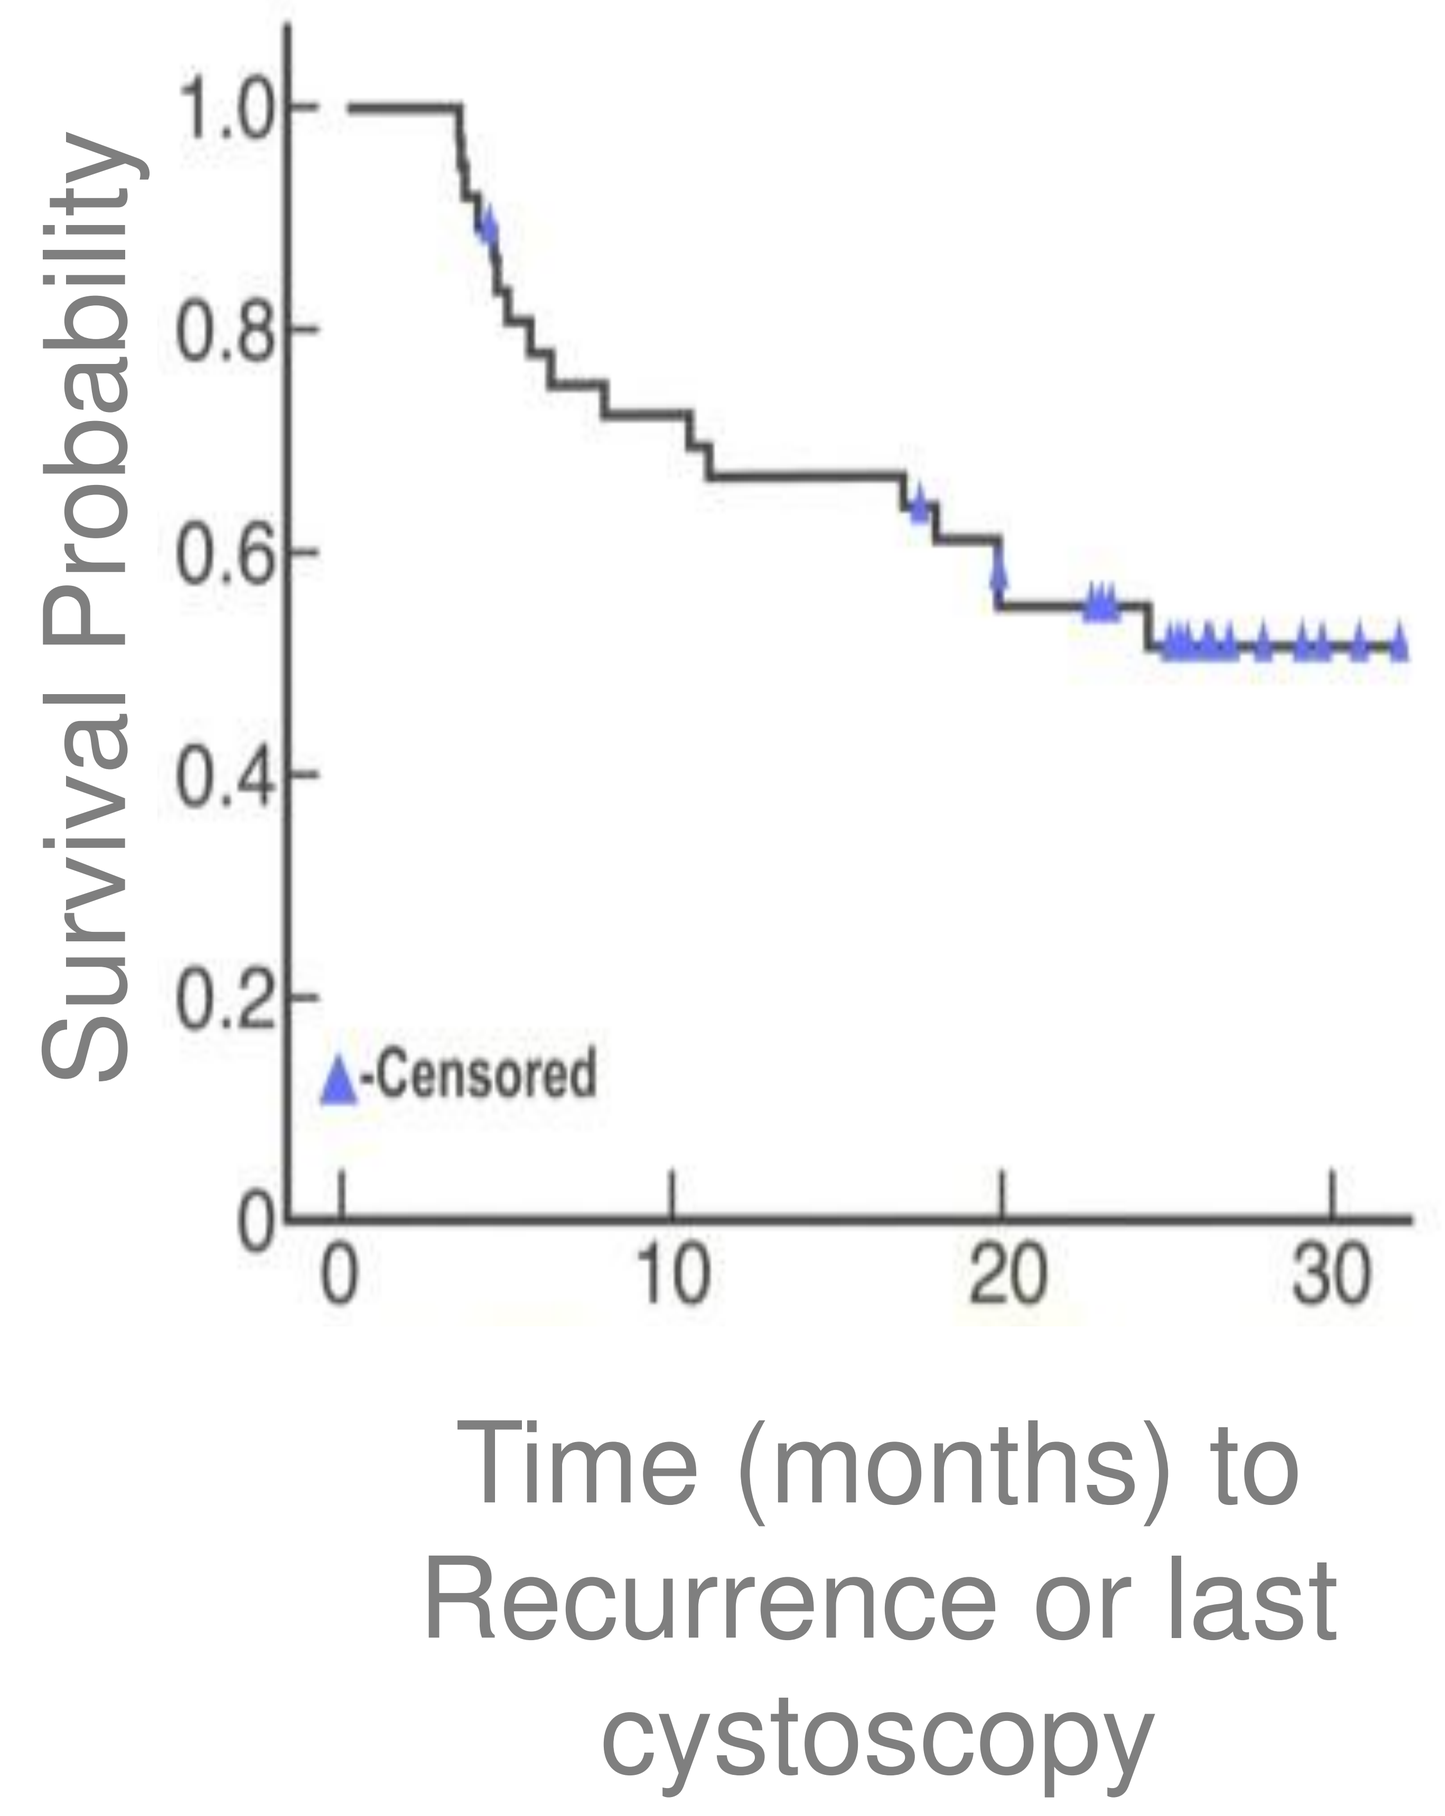
Survival Probability
1.0
0.8
0.6
0.4
0.2
0
-Censored
0
10
20
30
Time (months) to Recurrence or last cystoscopy

Conclusions:

- These data demonstrate that administration of BC-819 in combination with BCG is feasible and exhibits clinically meaningful activity in all three induction schedules.
- The safety profile suggests that BC-819 is well tolerated and does not add substantial toxicity to intravesical therapy.
- A phase 3 randomized trial of BC-819 and BCG in patients with NMIBC is planned.