PEG-HBCO MEDIATED CARBON MONOXIDE AND OXYGEN TRANSFER TO HYPOXIC RED BLOOD CELLS PREVENT, SLOW, AND/OR REVERSE SICKLING IN VITRO.

5351

Peter Buontempo, Cathy Buontempo, Ronald Jubin, and Abe Abuchowski,.

Prolong Pharmaceuticals, South Plainfield NJ

Background:

Background: In Sickle Cell disease (SCD), a single amino acid substitution in the beta globin chain converts HbA to sickle genotype HbS. This genetic change promotes HbS polymerization upon deoxygenation that can promote occlusion of small blood vessels that is often associated with increased blood viscosity, and circulatory inflammation. PEGylated-carboxyhemoglobin (PEG-COHb; SANGUINATE) was designed as a novel therapeutic agent to initially release carbon monoxide (CO) and then transfer oxygen (O2) to hypoxic tissue and cells. Delivery of either CO and/or O2to hypoxic, sickled red blood cells (RBCs) should return cells to a more normal cell morphology and help re-establish normal blood flow and rheology. PEG-COHb was shown to mediate transfer of either a CO or O2/CO mixture and restore normal morphology to hypoxic, sickled RBCs in vitro. Studies are now focused on the potential therapeutic implications of delaying or slowing sickling, which should maintain normal blood flow through hypoxic microvasculature. Unsickling is expected to be expedited by O2 transfer by PEG-COHb. To examine these potential therapeutic effects, current in vitro studies examined the effects of time and dose of PEG-COHb to not only reverse, but also prevent or delay sickling by transferring CO as well as expedite atmospheric O2 transfer to the sickled RBCs.

<u>Methods</u>: Reversal of sickling studies were conducted by deoxygenating RBCs from age matched healthy (control) and SCD volunteers followed by treatment with either PEG-COHb, fully oxygenated PEG-Hb (PEG-OHb) or PEG-BSA for 2 hours. For prevention of sickling studies, fully oxygenated RBC suspensions were treated with increasing amounts of PEG-COHb and then subjected to hypoxia for 3 hours. Time-dose effects were quantified by area under the curve (AUC) analysis. O2 transfer studies were conducted by treating hypoxic, sickled RBCs to increasing concentrations of PEG-COHb and raising the pO2 from 3.8mm to 40mm. In all studies, the fractions of CO-Hb, O2-Hb and reduced Hb were determined by co-oximetry and sickled RBCs were quantified by imaging flow cytometry of fixed RBC specimens.

KEY:

PEG-COHb: PEGylated CarboxyHemoglobin, bovine

PEG-OHb: PEGylated OxyHemoglobin, bovine

PEG-Alb: PEGylated Albumin, bovine

PEG-COHb-Mediated Transfer of CO and O_2 Reverses and Prevents Sickling

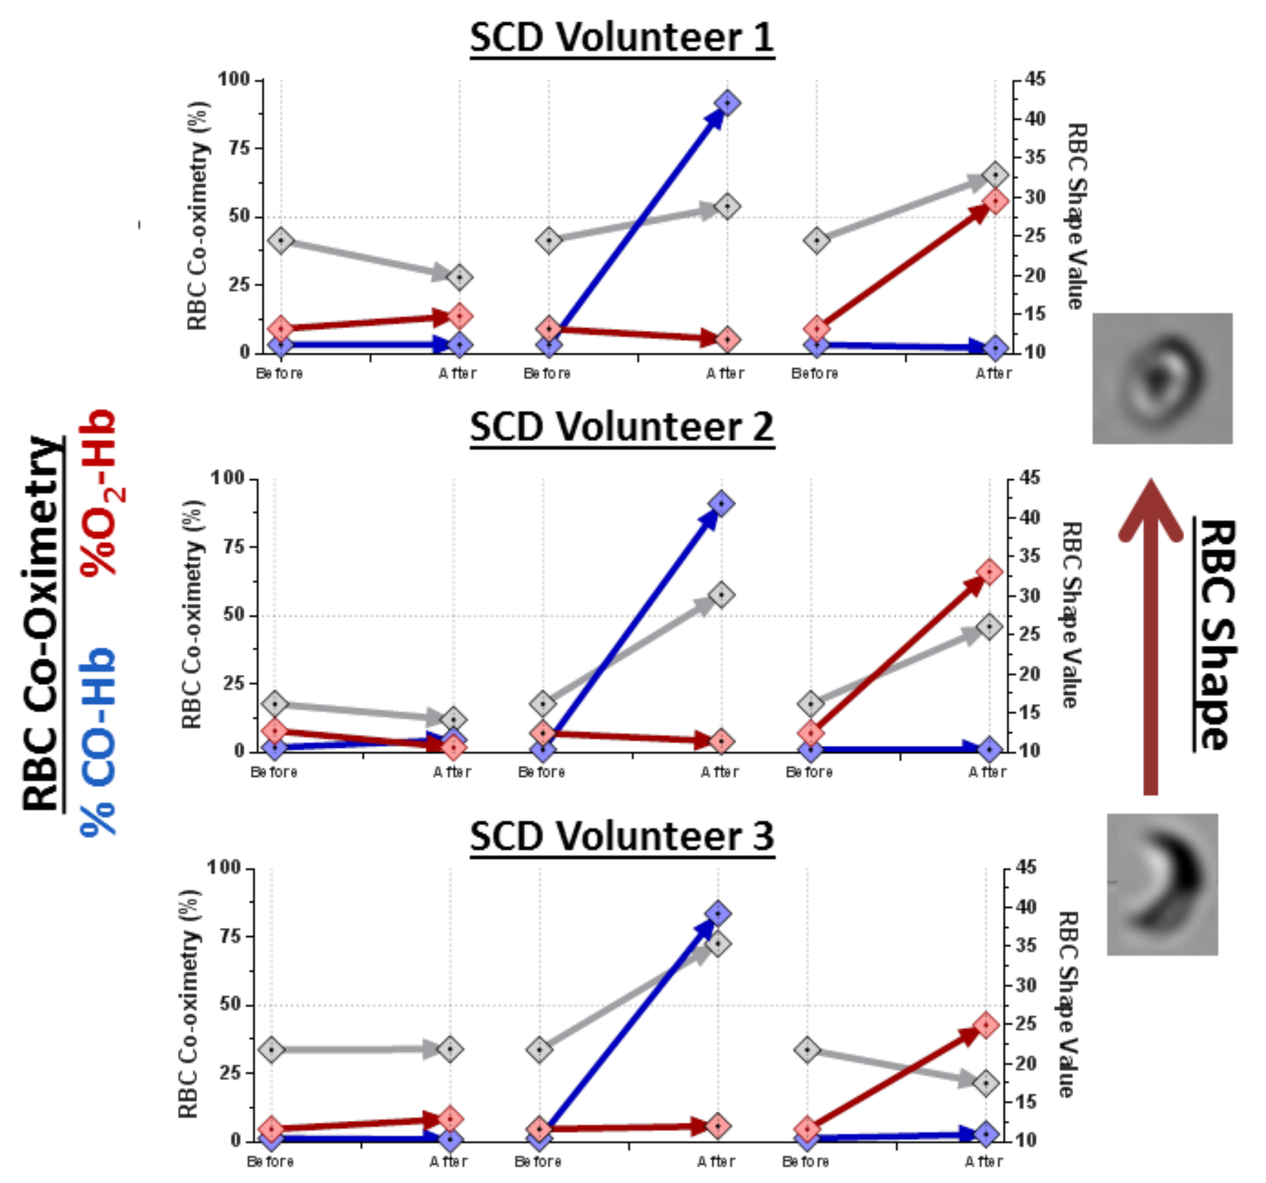
SCD Volunteer 1
SCD Volunteer 2
SCD Volunteer 3
RBC Co-Oximetry
% CO-Hb
%O2-Hb
RBC Shape

RBC from SCD and healthy volunteers were incubated at 3.8 mm pO2 for 3 hours prior to treatment with PEG-COHb or PEG-OHb. Sample was fixed for imaging cytometry and the percentage of carboxy- and oxyhemglobin determined.

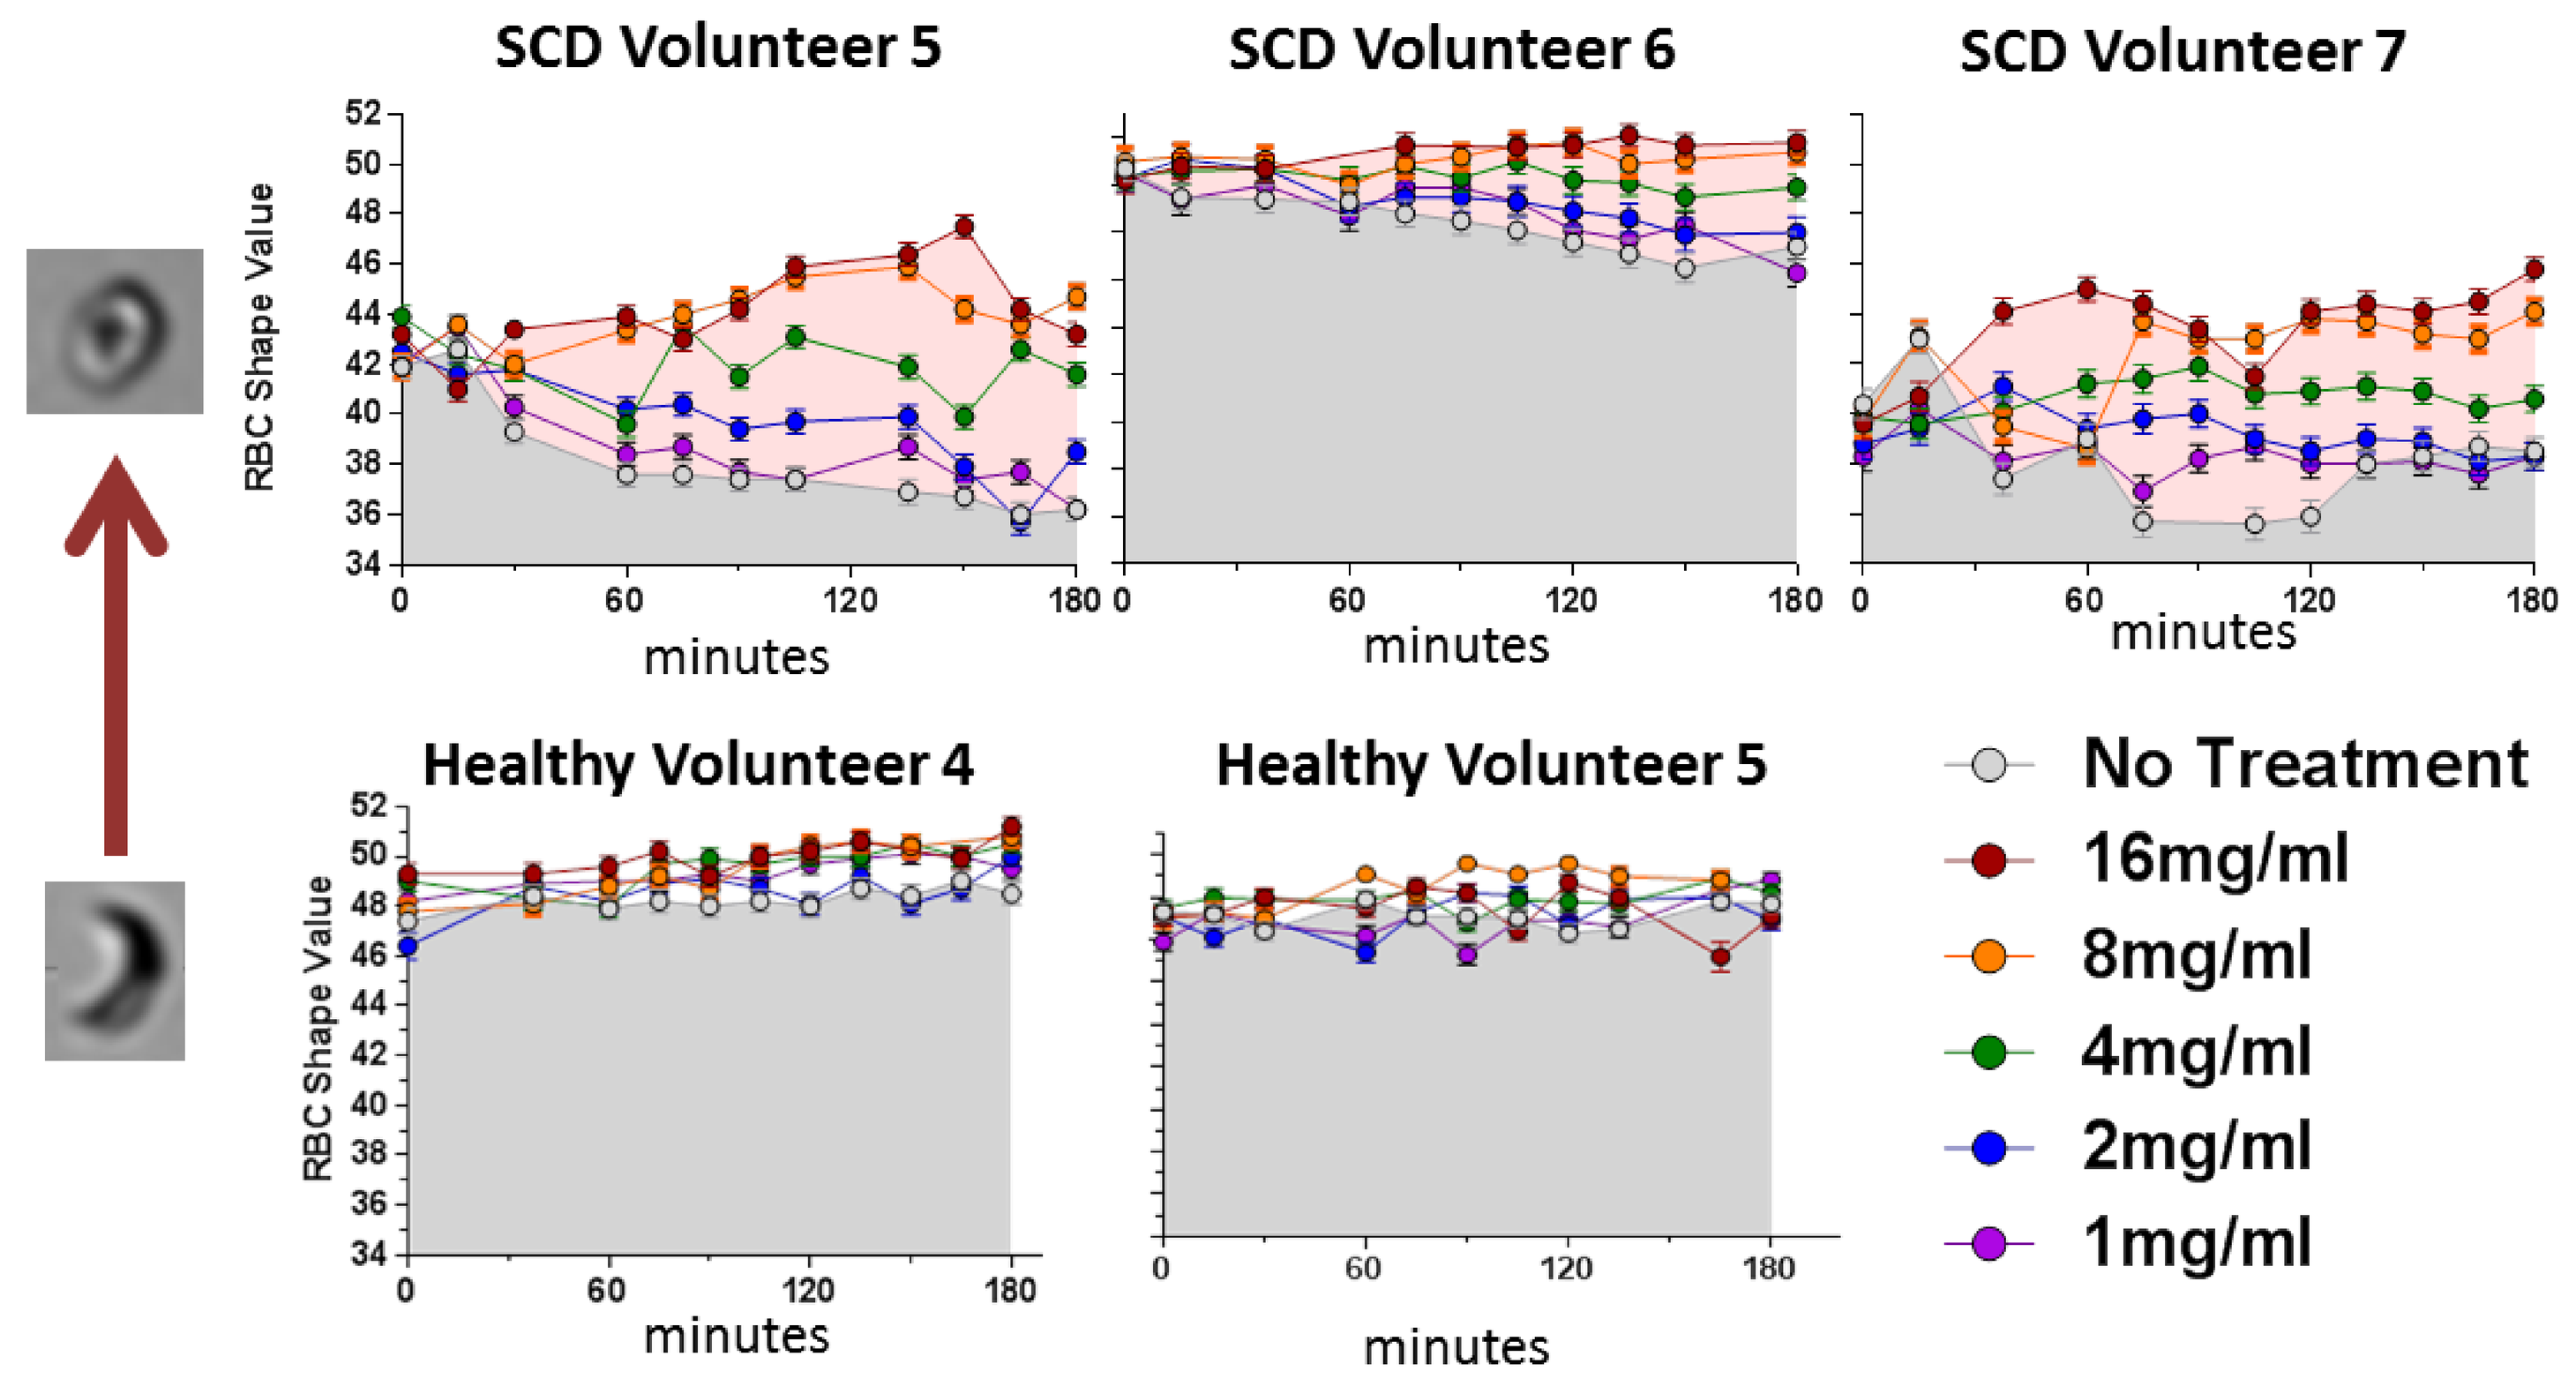
SCD Volunteer 5
SCD Volunteer 6
SCD Volunteer 7
Healthy Volunteer 4
Healthy Volunteer 5
RBC Shape Value
minutes
No Treatment
16mg/ml
8mg/ml
4mg/ml
2mg/ml
1mg/ml

Mean red blood cell shape value were plotted as a function of time. Gray and red shaded areas represent the area under the response curve (AUC) of the no treatment and 8 mg per mL

PEG-COHb-Mediated O_2 Unsickling is More Rapid than CO

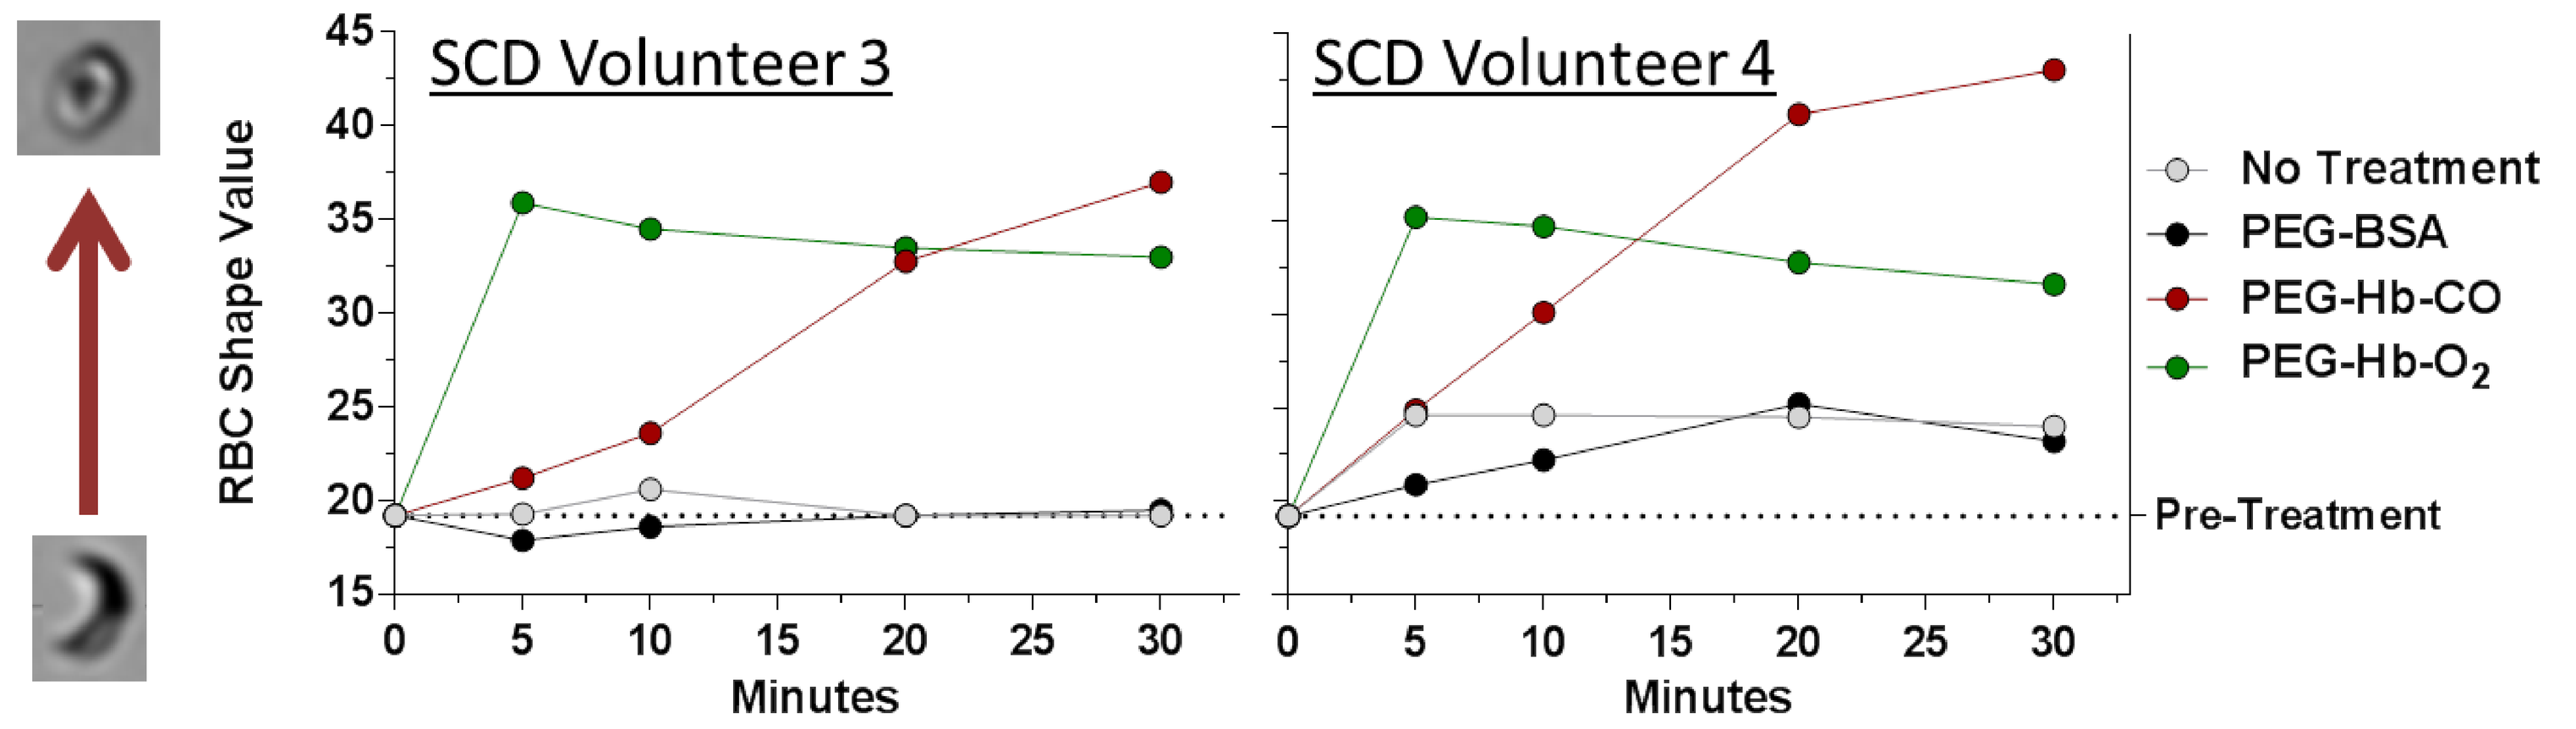
SCD Volunteer 3
SCD Volunteer 4
RBC Shape Value
Minutes
No Treatment
PEG-BSA
PEG-Hb-CO
PEG-Hb-O2
Pre-Treatment

Sickled RBCs red blood cells from SCD volunteers were treated with bovine PEG-COHb or PEG-OHb and fixed at recurrent intervals and subjected to imaging cytometry.

PEG-COHb speeds O_2 mediated unsickling

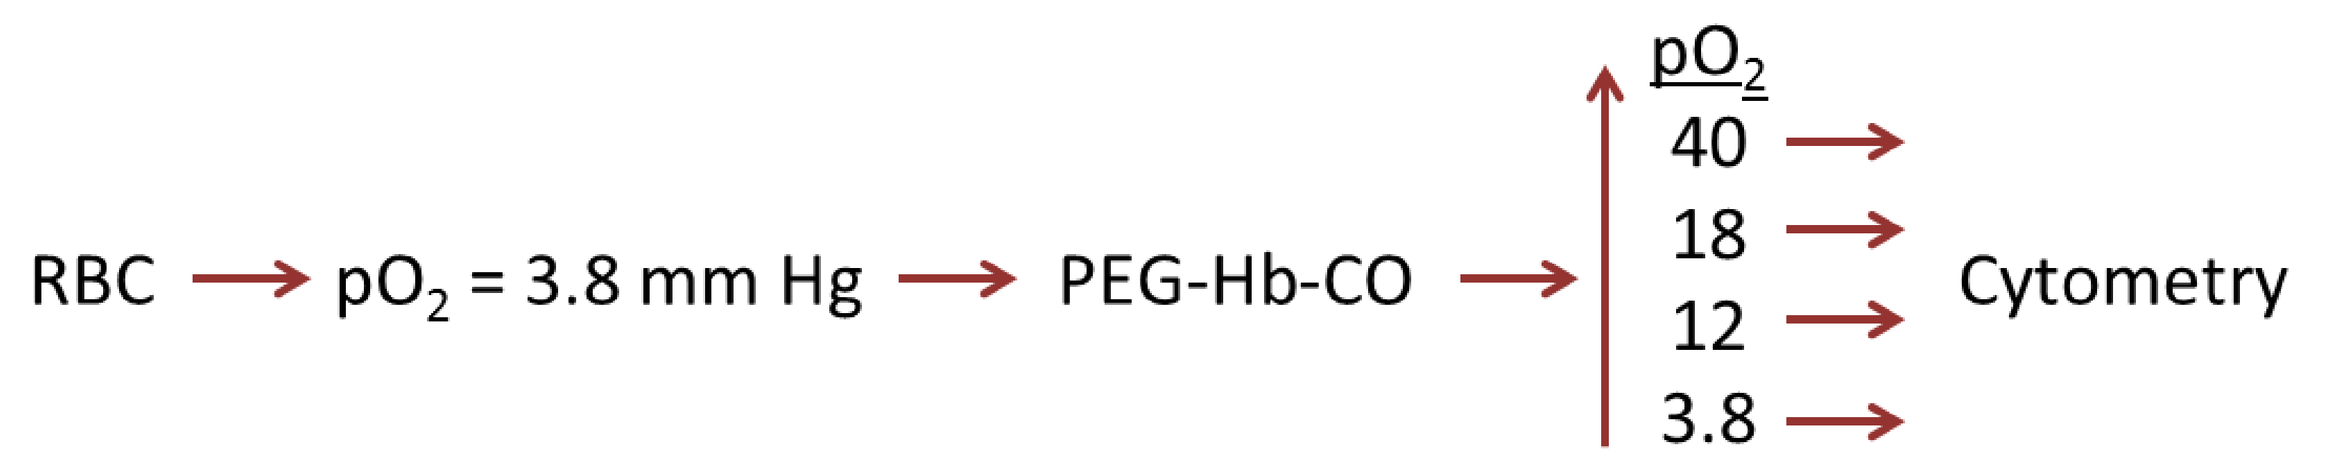
RBC
pO2 = 3.8 mm Hg
PEG-Hb-CO
pO2
40
18
12
3.8
Cytometry

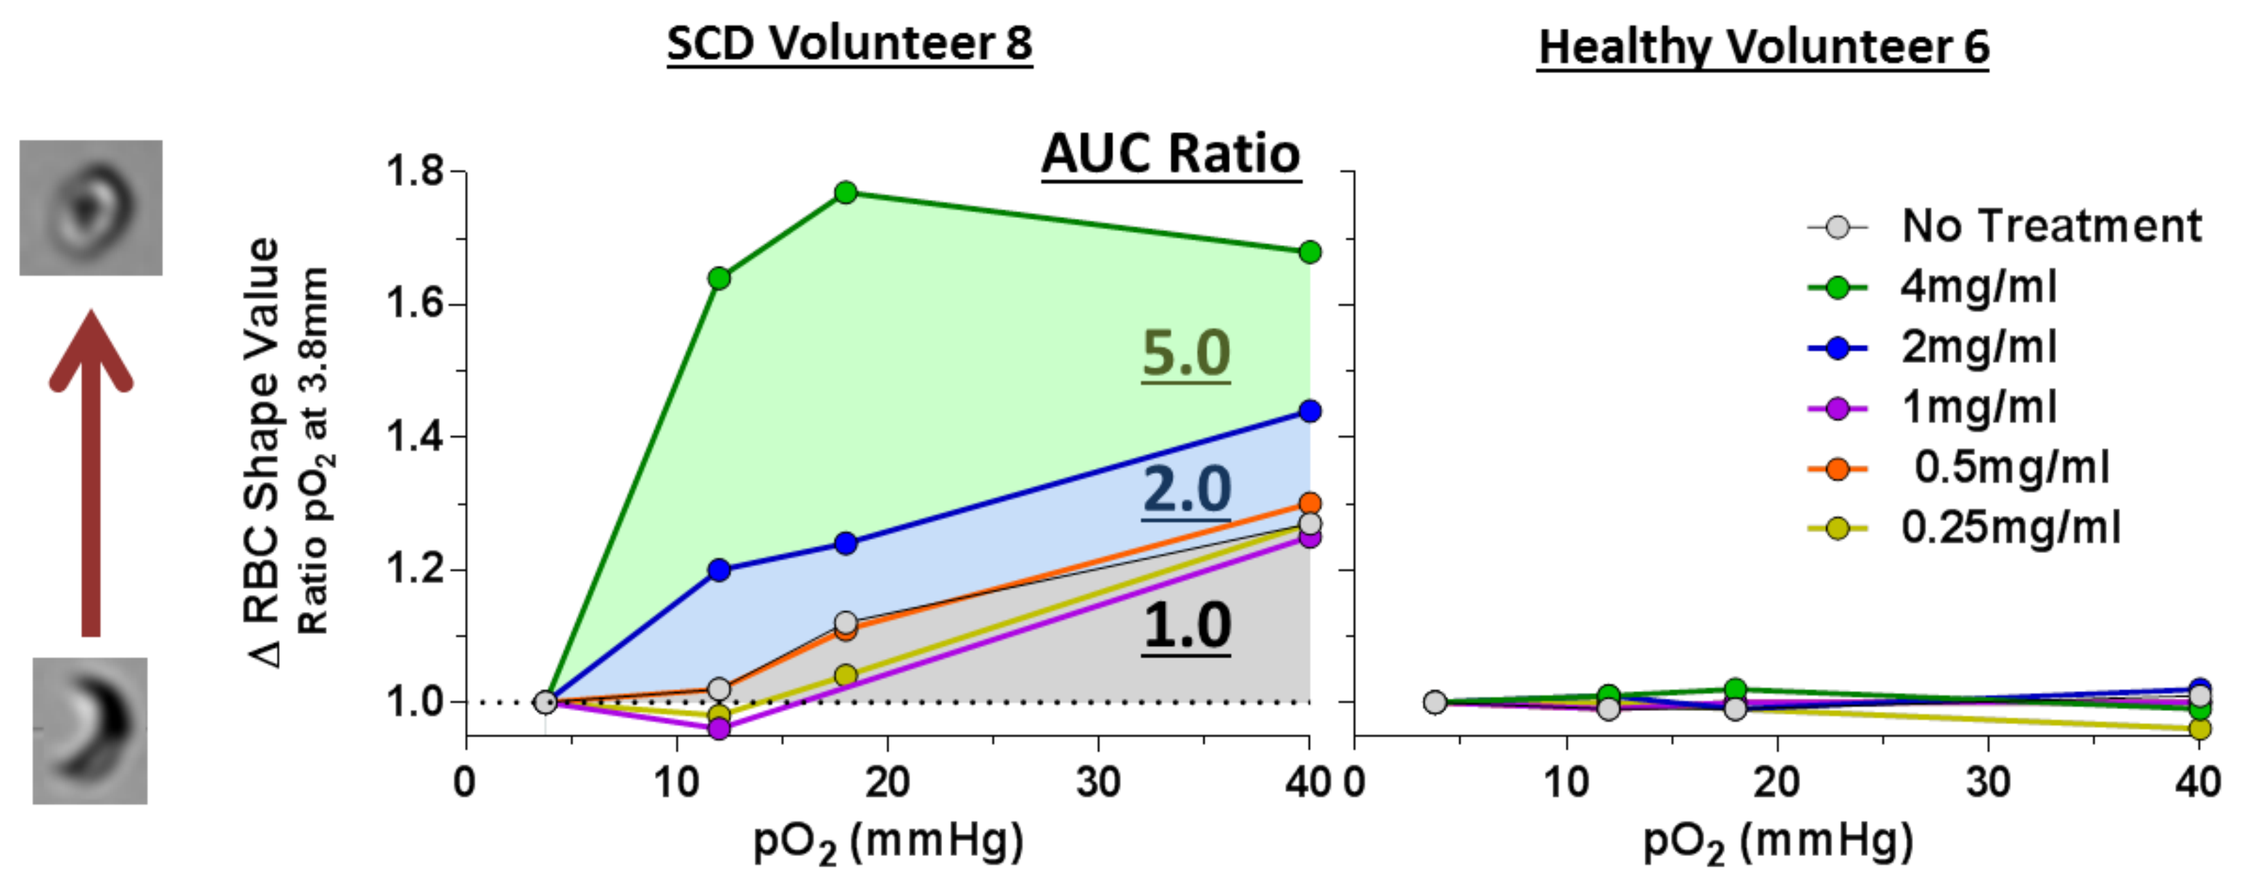
SCD Volunteer 8
Healthy Volunteer 6
AUC Ratio
5.0
2.0
1.0
Δ RBC Shape Value
Ratio pO2 at 3.8mm
pO2 (mmHg)
No Treatment
4mg/ml
2mg/ml
1mg/ml
0.5mg/ml
0.25mg/ml

RBC from SCD and healthy volunteers were treated with PEG-COHb after incubating at 3.8 mm pO2. O_2 was raised in stages to 12, 18 & 40 mmHg & samples taken to quantify shape change. Data is expressed as fractional change of RBC shape at 3.8 mmHg as a function of mmHg pO2 to calculate AUC. Green, blue and gray shaded areas represent AUC values of 4, 2, and 0 mg/mL, respectively. Ratio of AUC values to untreated control are shown.

Summary:

RBCs from patients with SCD undergo a conformational shift upon deoxygenation resulting in HbS polymerization and morphological changes of the RBCs. The occlusive and fragile properties of sickled RBCs are responsible for the development of the numerous comorbidities associated with SCD. It is only when the fraction of oxygenated or carboxylated HbS reaches a sufficient level that reversion to normal cell morphology occurs. These experiments showed a concentration and time-dependent effect of PEG-COHb ability to deliver both O2 and CO to sickled RBC. These data suggested that PEG-COHb is a promising gas transfer agent that has the potential to improve sickle cell morphology by reversing sickling; the underlying pathology of sickle cell disease co-morbidities.

For More Info Please Contact: www.prolongpharma.com

The Conflict of Interest disclosure forms for above authors have been satisfied.